DTG + 3TC IN GEMINI-1 & -2: HIV-1 REPLICATION AT <50 C/ML AND VL 'BLIPS' THROUGH 144 WEEKS

Mark Underwood,[1] **Rimgaile Urbaityte**,[2] **Ruolan Wang**,[1] **Joe Horton**,[3] **Linshan Yuan**,[4] **Brian Wynne**,[1] **Justin Koteff**,[1] **Jean van Wyk**,[5] **Choy Man**,[1] **Jörg Sievers**[5]

[1]ViiV Healthcare, Research Triangle Park, NC, USA; [2]GlaxoSmithKline, Brentford, UK; [3]Parexel, Durham, NC, USA; [4]GlaxoSmithKline, Collegeville, PA, USA; [5]ViiV Healthcare, Brentford, UK

PEB163

Introduction

- The GEMINI-1 & -2 studies in treatment-naive adults (Figure 1) showed DTG + 3TC was non-inferior to DTG + TDF/FTC by FDA Snapshot (HIV-1 RNA <50 c/mL) at Week 144.[1]
 - HIV-1 RNA <50 c/mL was also comparable across baseline subgroups including HIV-1 RNA > or ≤100,000 c/mL and CD4+ cell count > or ≤200 cells/mm^3.
- Abbott's RealTime HIV-1 assay provides quantitative viral load (VL) from 40 to 10,000,000 c/mL, and qualitative target detected (TD) or target not detected (TND) for VL <40 c/mL.
- In this post hoc analysis, we assessed very-low-level viremia by TND and TD, and low-level quantitative viral replication including 'blips' through Week 144 overall, and by baseline stratification subgroups.

Figure 1. GEMINI-1 and GEMINI-2 Study Design

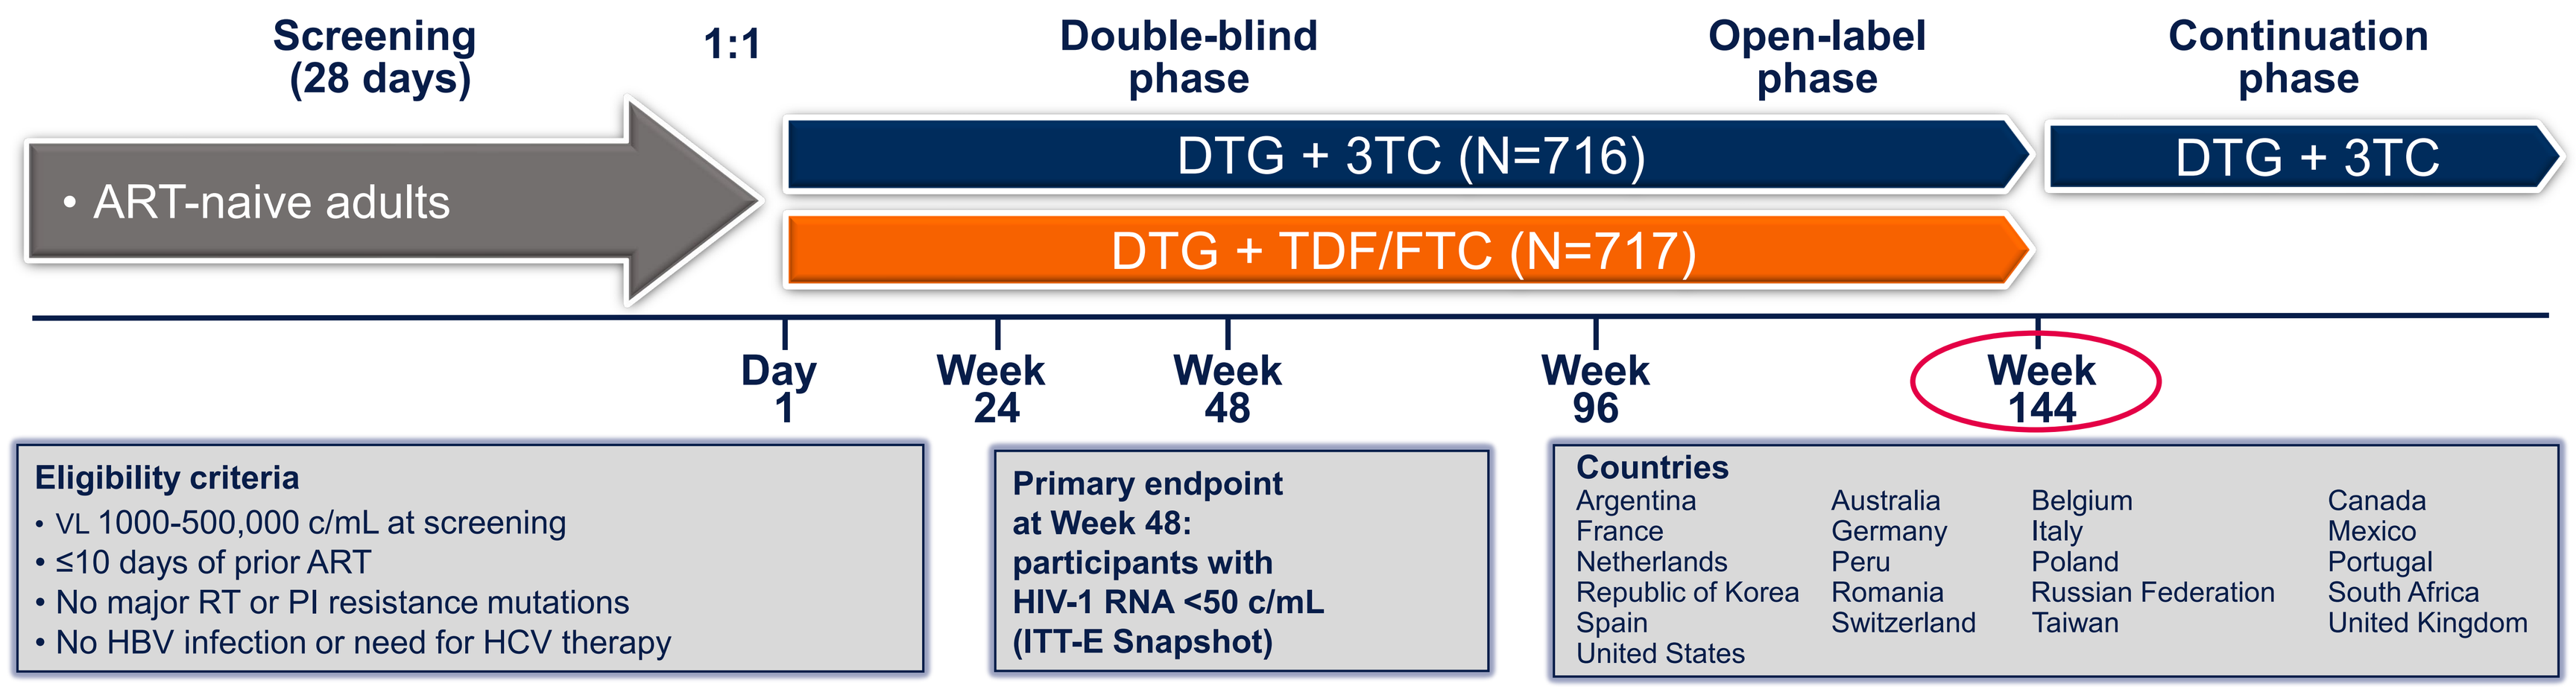
Screening (28 days)
1:1
Double-blind phase
Open-label phase
Continuation phase
• ART-naive adults
DTG + 3TC (N=716)
DTG + TDF/FTC (N=717)
DTG + 3TC
Day 1
Week 24
Week 48
Week 96
Week 144
Eligibility criteria
• VL 1000-500,000 c/mL at screening
• ≤10 days of prior ART
• No major RT or PI resistance mutations
• No HBV infection or need for HCV therapy
Primary endpoint at Week 48: participants with HIV-1 RNA <50 c/mL (ITT-E Snapshot)
Countries
Argentina, France, Netherlands, Republic of Korea, Spain, United States
Australia, Germany, Peru, Romania, Switzerland
Belgium, Italy, Poland, Russian Federation, Taiwan
Canada, Mexico, Portugal, South Africa, United Kingdom

TND

Methods and Assessments

- The proportion of participants with VL <40 c/mL and TND status throughout Week 144 is presented based on Snapshot analysis via a Cochran-Mantel-Haenszel stratified analysis, adjusting for the following baseline stratification factors:
- Plasma HIV-1 RNA (≤100,000 c/mL vs >100,000 c/mL) and CD4+ cell count (≤200 cells/mm^3 vs >200 cells/mm^3) and Study (204861 vs 205543).
- Participant subgroups assessed included the ITT-E population and an Observed subpopulation (ITT-E participants with VL <50 c/mL at Week 144).
- Participants with VL ≥50 c/mL were assigned to mutually exclusive VL groups as previously presented.[2] Those with no VL ≥200 c/mL could have a single VL between 50 to <200 c/mL with adjacent values <50 c/mL ('blip' definition) or ≥ two consecutive VLs between 50 to <200 c/mL. Those with ≥1 VL ≥200 c/mL could meet SVW criteria with a single occurrence or meet CVW criteria with consecutive VL ≥200 c/mL.

Figure 2. Participant Proportions* With TND or TD Were Similar Across Arms From Week 4 Through Week 144, ITT-E Population

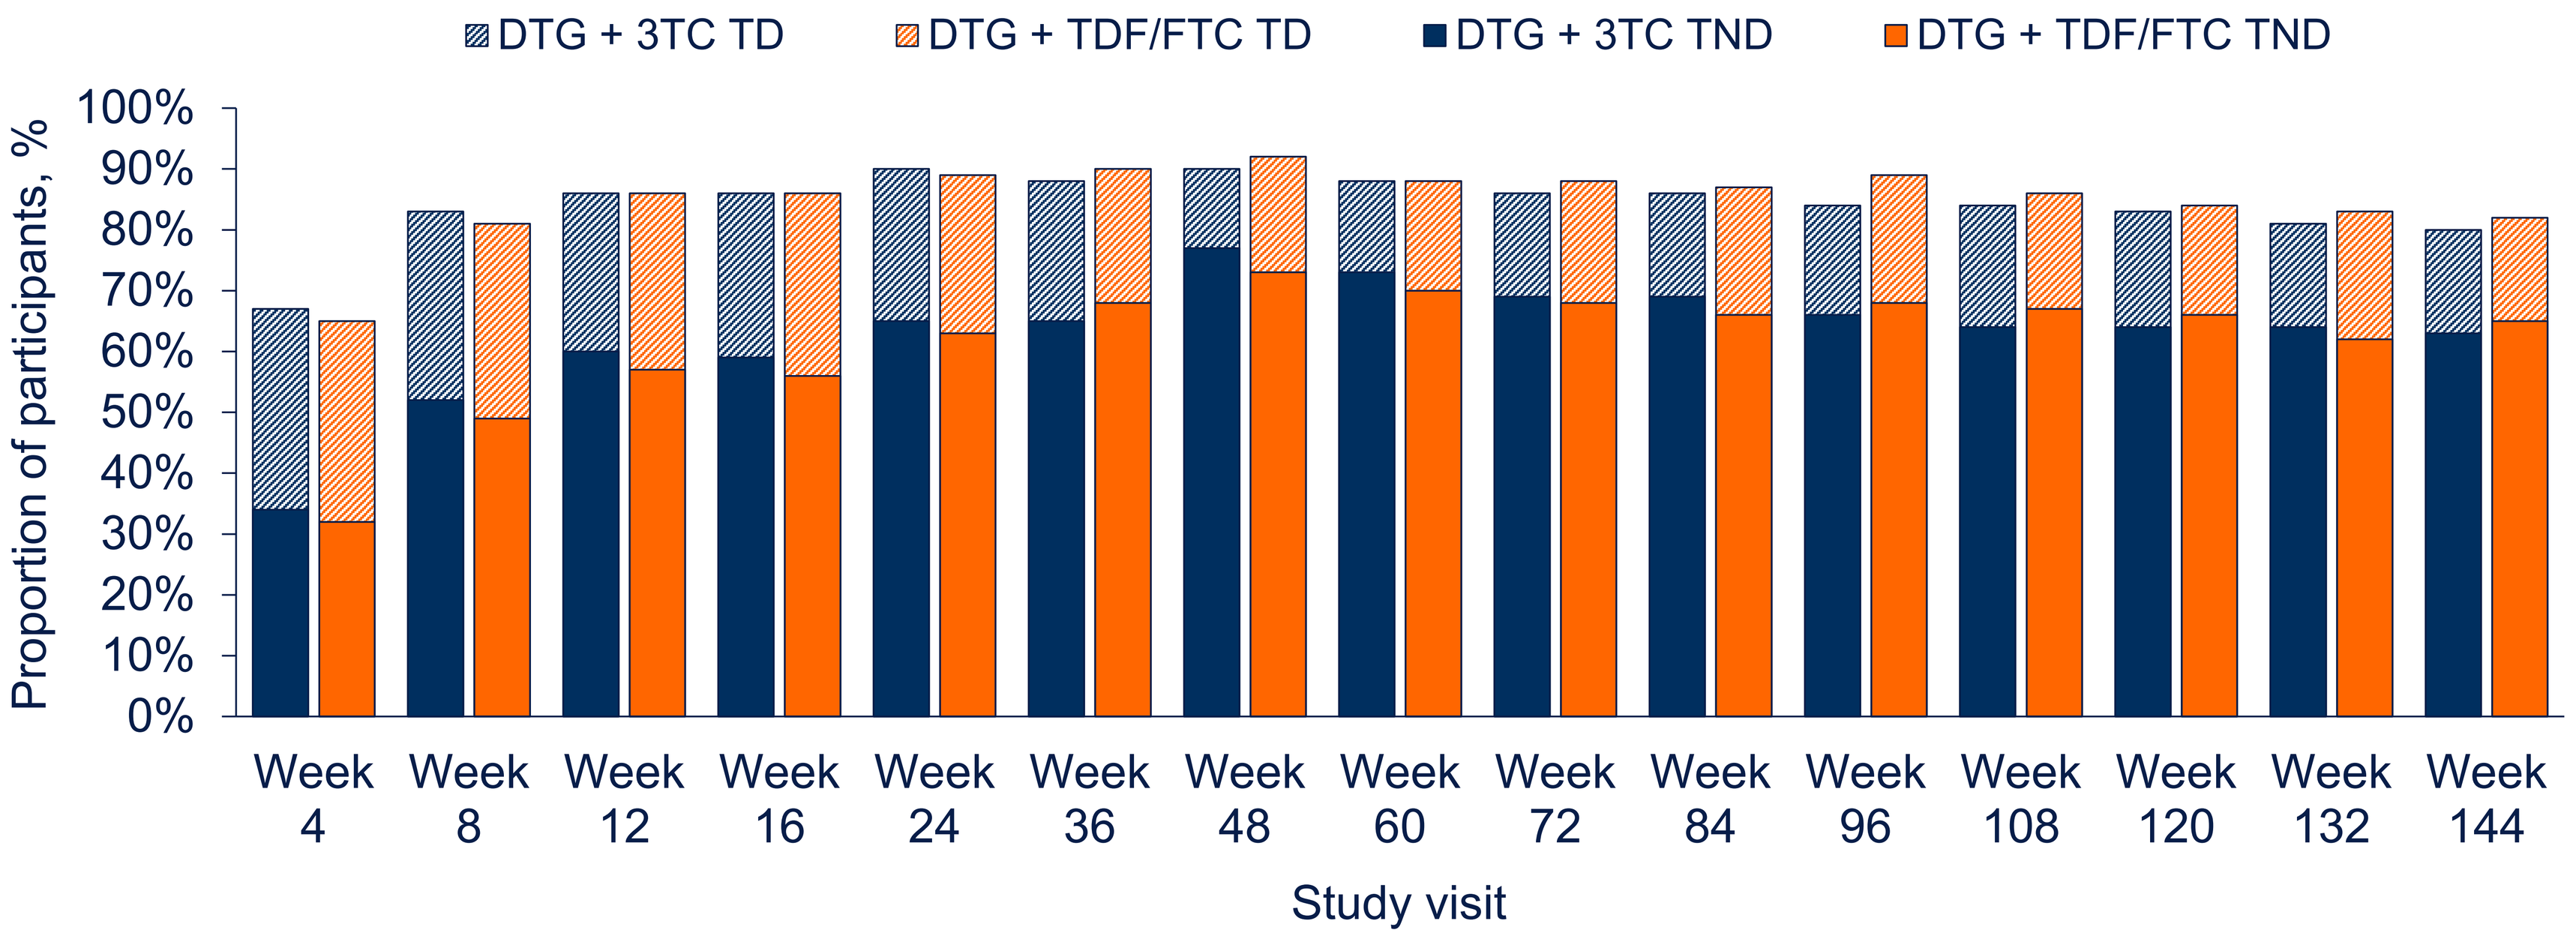
DTG + 3TC TD
DTG + TDF/FTC TD
DTG + 3TC TND
DTG + TDF/FTC TND
Proportion of participants, %
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
Week 4
Week 8
Week 12
Week 16
Week 24
Week 36
Week 48
Week 60
Week 72
Week 84
Week 96
Week 108
Week 120
Week 132
Week 144
Study visit

*Percentages calculated for DTG + 3TC and DTG + TDF/FTC in Fig. 2 used, respectively, N=716 and N=717 ITT-E population as denominators.

- Proportions with TND trended upward through ~Week 48 and were similar between arms at all visits.
- The proportions that had highest TD occurred in earlier visits and decreased to stabilize around Week 48.

Figure 3. Proportions* of Participants With TND by BL Subgroups Were Similar Across Arms for ITT-E (Overall) and Observed (OBS) Populations at Week 144

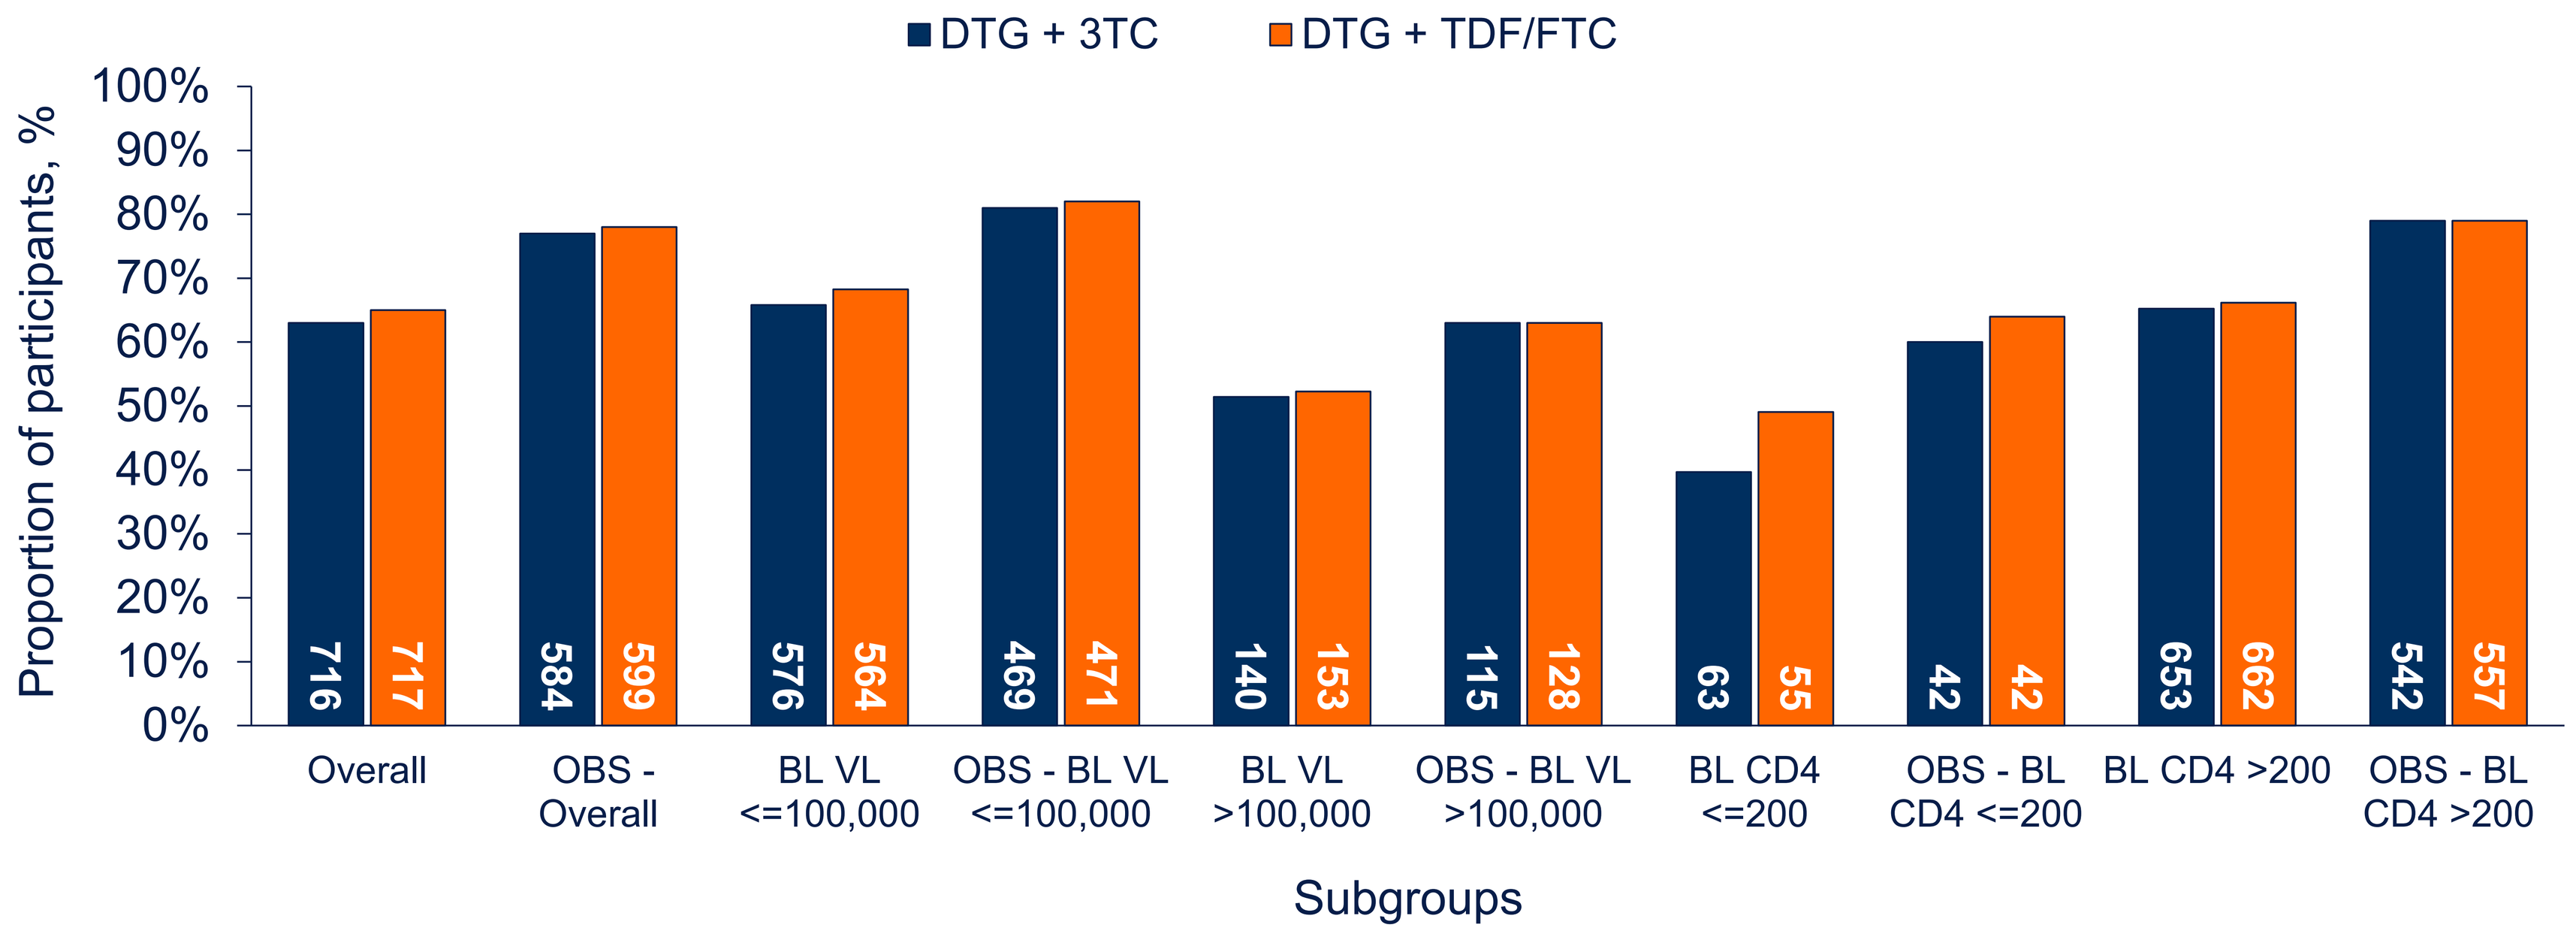
DTG + 3TC
DTG + TDF/FTC
Proportion of participants, %
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
716
717
584
599
576
564
469
471
140
153
115
128
63
55
42
42
653
662
542
557
Overall
OBS - Overall
BL VL <=100,000
OBS - BL VL <=100,000
BL VL >100,000
OBS - BL VL >100,000
BL CD4 <=200
OBS - BL CD4 <=200
BL CD4 >200
OBS - BL CD4 >200
Subgroups

*Percentages calculated for DTG + 3TC and DTG + TDF/FTC in Fig. 3 used ITT-E Overall or Observed and HIV-1 RNA and CD4 subgroups as denominators, which are in white overlaid on vertical bars.

- At Week 144, there were similar proportions of ITT-E Overall participants with TND receiving DTG + 3TC vs DTG + TDF/FTC by Snapshot (63% [451/716] vs 65% [465/717]), or for Observed (OBS) population (77% [451/584] vs 78% [465/599]).
- Proportions with TND within subgroups were generally similar between arms.
- For the ITT-E Overall group, the largest difference was seen with BL CD4 ≤200.
 - Proportion with TND was 40% for the DTG + 3TC arm compared with 49% for the DTG + TDF/FTC arm, though the Ns were very small, at 25/63 and 27/55, respectively.
 - For the Observed population, the difference in proportions with TND seen with BL CD4 ≤200 was smaller at 60% vs 64%, with Ns of 25/42 and 27/42, respectively.

Discussion

- Two-drug regimens (2DRs) reduce the number of drugs for PLWH who need lifelong ART.[3]
- The implications of low-level qualitative HIV-1 replication (TD and TND) remain exploratory, and without known or apparent clinical consequences. Previously reported assessments of very-low-level TND and TD for qualitative HIV-1 RNA <40 c/mL for the GEMINI studies at Week 48[4] showed similar frequency of participants across arms with undetectable HIV-1 RNA, although the median time to undetectable HIV-1 RNA was numerically shorter for DTG + 3TC participants vs DTG + TDF/FTC participants with BL VLs >100,000 c/mL.
- Low-level single HIV-1 VL elevations known as 'blips' (HIV-1 RNA of 50-200 c/mL preceded and followed by HIV-1 RNA <50 c/mL) are generally considered as clinically not impactful.[5] As previously reported,[2] 'blip' assessments for the GEMINI studies at Week 48 showed proportion with 'blips' through 48 weeks was overall similar between the DTG + 3TC and DTG + TDF/FTC arms, though a higher percentage of 'blips' occurred in participants receiving DTG + TDF/FTC than DTG + 3TC if BL VL was >100,000 c/mL.

'Blips'

Methods and Assessments

- 'Blips' by visit were assessed from Day 1 after VL suppression to <50 c/mL, and from Weeks 48 through 144. Percentages are based on number of 'blips' and available VLs in the analysis window. Multiple VLs from the same visit week are included, if applicable. Participants who never suppressed, or VL records before and including the first suppression <50 c/mL, are excluded.

Figure 4. Participant Proportions* With 'Blips' Were Similar Across Arms by Visit Through Week 144, ITT-E Population

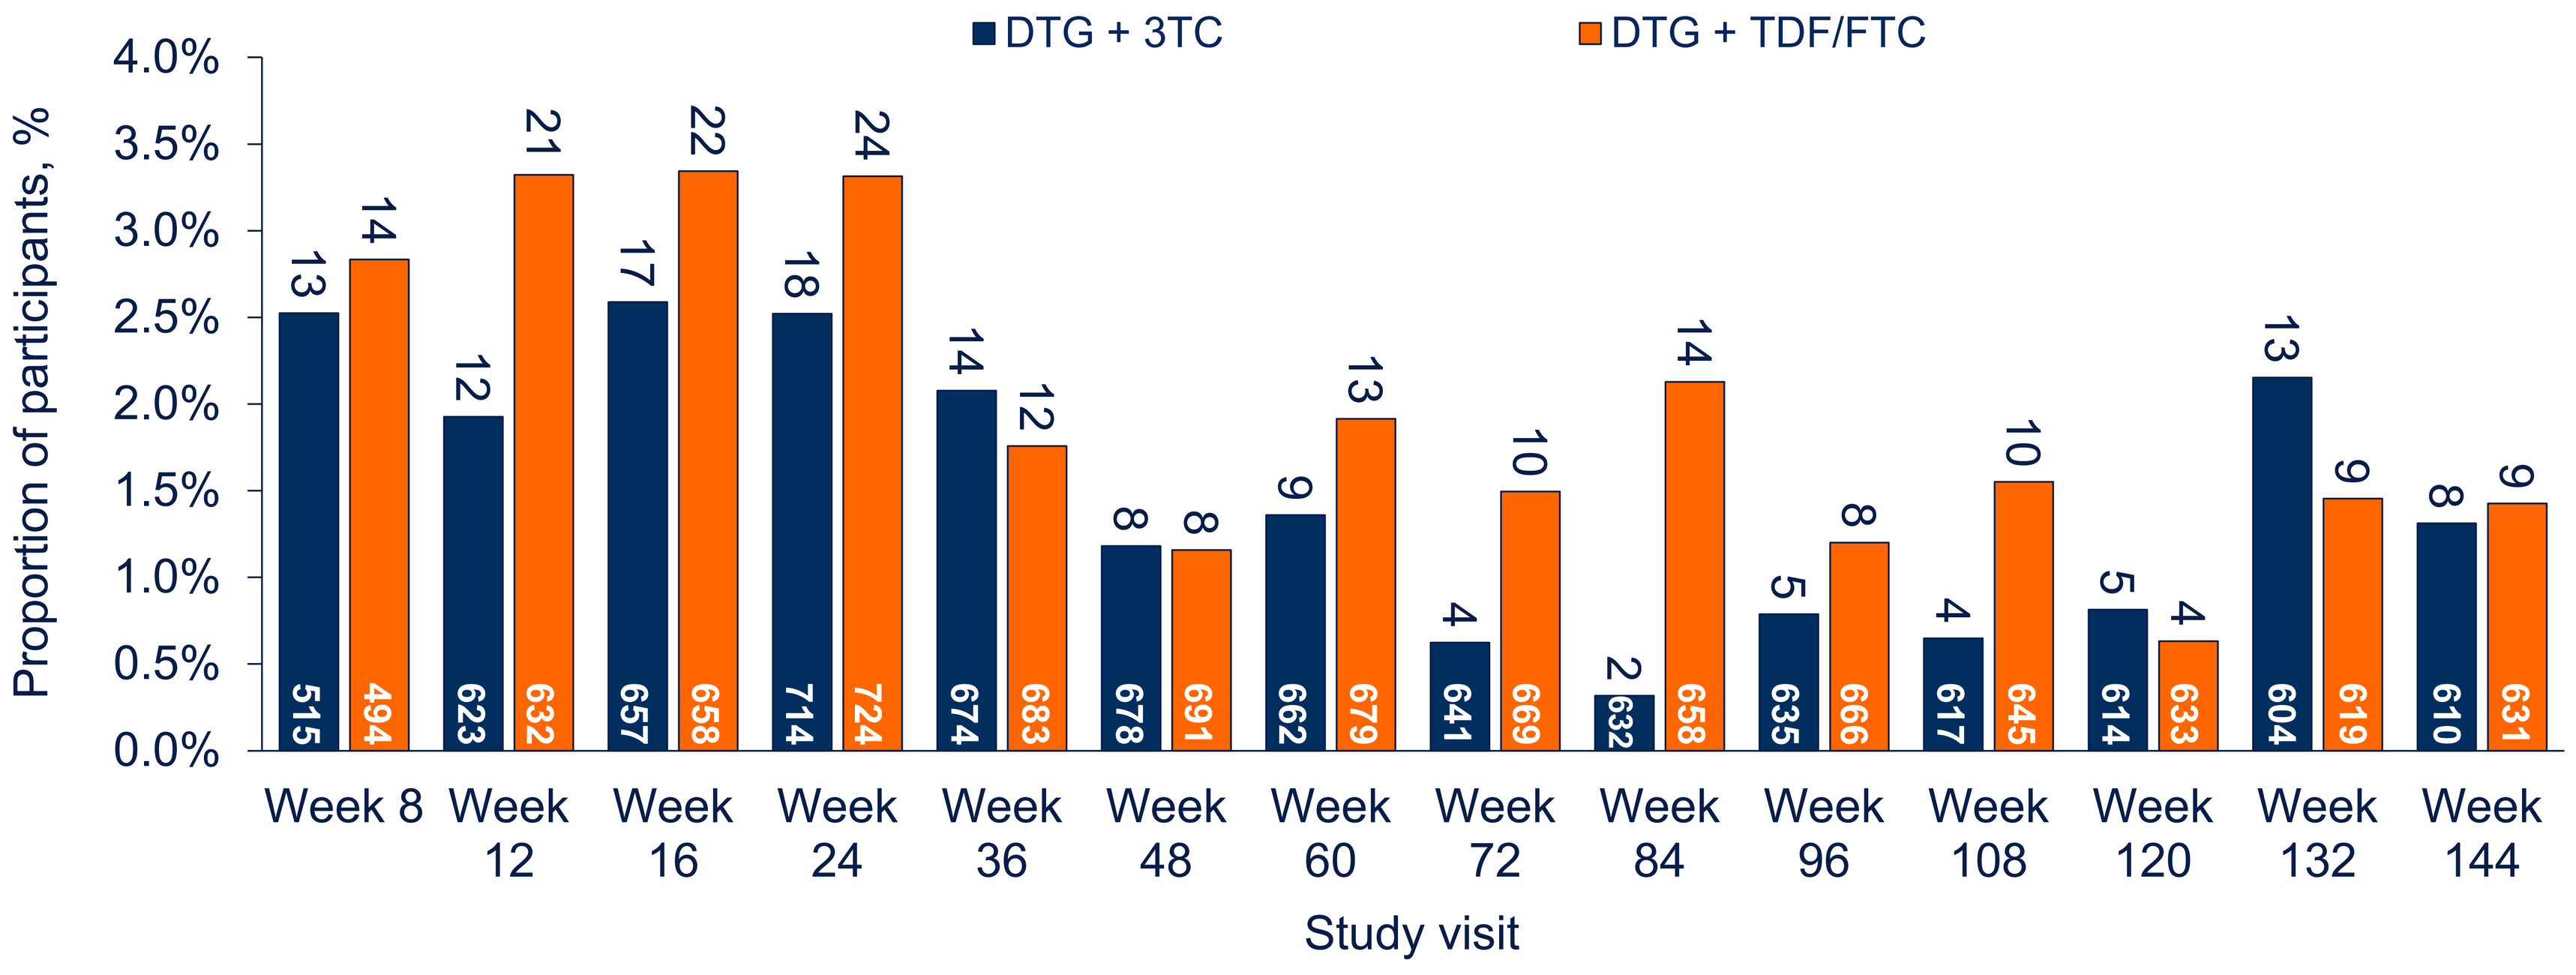
DTG + 3TC
DTG + TDF/FTC
Proportion of participants, %
4.0%
3.5%
3.0%
2.5%
2.0%
1.5%
1.0%
0.5%
0.0%
13 14
515 494
12 21
623 632
17 22
657 658
18 24
714 724
14 12
674 683
8 8
678 691
9 13
662 679
4 10
641 669
2 14
632 658
5 8
635 666
4 10
617 645
5 4
614 633
13 9
604 619
8 9
610 631
Week 8
Week 12
Week 16
Week 24
Week 36
Week 48
Week 60
Week 72
Week 84
Week 96
Week 108
Week 120
Week 132
Week 144
Study visit

*The denominator is in white overlaid on the vertical bars and is the total number of observations from all participants with data for the specified visit window. Numbers on tops of bars are # of 'blips' at given week visits. Note that individual can have had more than one 'blip' (see Table 1).

- 'Blip' proportions were higher early and decreased around Weeks 36 to 48.
- The proportion of 'blips' was generally numerically lower for DTG + 3TC but similar across arms through Week 144.

Figure 5. Participant Proportions* With ≥1 'Blip' From Day 1 to Week 144 or Week 48 to Week 144 by BL Subgroups Were Generally Similar Across Arms

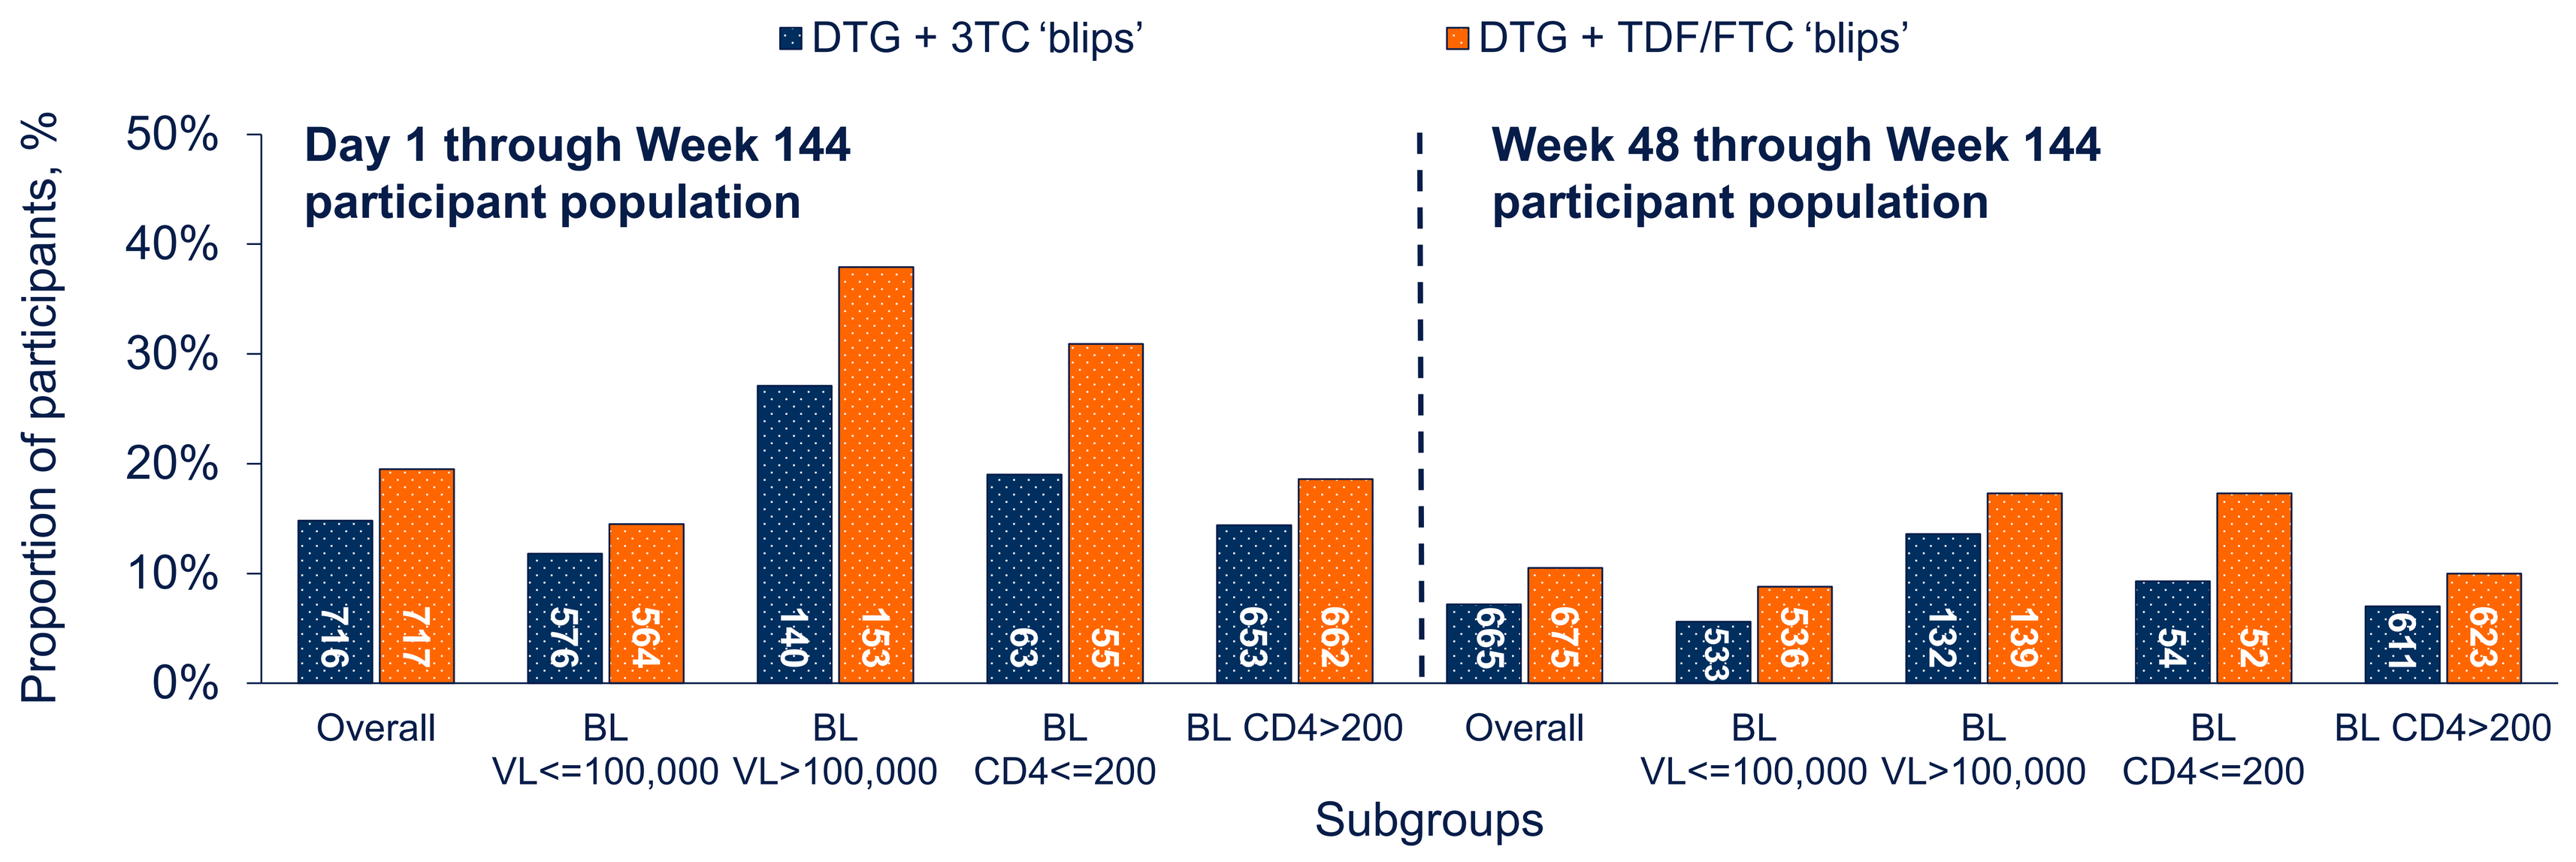
DTG + 3TC 'blips'
DTG + TDF/FTC 'blips'
Proportion of participants, %
50%
40%
30%
20%
10%
0%
Day 1 through Week 144 participant population
Week 48 through Week 144 participant population
716 717
576 564
140 153
63 55
653 662
665 675
533 536
132 139
54 52
611 623
Overall
BL VL<=100,000
BL VL>100,000
BL CD4<=200
BL CD4>200
Overall
BL VL<=100,000
BL VL>100,000
BL CD4<=200
BL CD4>200
Subgroups

*Percentages calculated for DTG + 3TC and DTG + TDF/FTC in Fig. 5 use ITT-E or subgroup Ns (by BL VL or CD4) as denominators shown in white on columns.

- There was a broadly lower 'blip' frequency from Week 48 through Week 144, ie, after participants had suppressed.
- The highest 'blip' proportion was observed for the Day 1 to Week 144 category in DTG + TDF/FTC participants with BL VL >100,000 c/mL or CD4+ ≤200 cells/mm^3.

Table 1. Participants With Different Numbers of 'Blips' From Day 1 to Week 144

'Blips' per participant	DTG + 3TC (%)	DTG + TDF/FTC (%)
1	83 (11.6%)	111 (15.5%)
2	20 (2.8%)	23 (3.2%)
3	3 (0.4%)	4 (0.6%)
4	0	1 (0.1%)
5	0	1 (0.1%)
Total	**106 (14.6%)**	**140 (19.5%)**

*Percentages calculated for DTG + 3TC and DTG + TDF/FTC, respectively, used N=716 and N=717 ITT-E population as denominators.

- Most participants with 'blips' had only 1, and this number was lower in the DTG + 3TC arm.
- No 'blips' were observed for participants meeting CVW criteria in either arm.

Conclusions

- Proportions of participants with TND were similar through Week 144 in the DTG + 3TC and DTG + TDF/FTC arms.
 - For participants with BL CD4+ cell count ≤200 cells/mm^3, the difference in TND proportion for DTG + 3TC vs DTG + TDF/FTC was smaller for the Observed population compared with the ITT-E population.
- The frequency of 'blips' through Week 144 was generally similar across arms when assessed early from Day 1 or from Week 48.
- These data continue to demonstrate the efficacy, potency, and durability of the DTG + 3TC 2DR in treatment-naive adults.

Acknowledgments: This study was funded by ViiV Healthcare. We thank everyone who has contributed to the success of these studies, including all study participants and their families; the GEMINI-1 and GEMINI-2 clinical investigators and their staff; and the ViiV Healthcare, PPD, Parexel, and GSK teams. Editorial assistance and graphic design support for this presentation were provided under the direction of the authors by Sydney Stocks and Dominick de la Guardia, and MedThink SciCom, and funded by ViiV Healthcare.

References: **1.** Cahn et al. HIV Glasgow 2020; Virtual. Poster P018. **2.** Underwood et al. IAS 2019; Mexico City, Mexico. Poster MOPEB231. **3.** Kelly et al. *Drugs.* 2016;76:523-531. **4.** Underwood et al. CROI 2019; Seattle, WA. Abstract 490. **5.** Panel on Antiretroviral Guidelines for Adults and Adolescents. https://clinicalinfo.hiv.gov/sites/default/files/guidelines/documents/AdultandAdolescentGL.pdf. Accessed June 16, 2021. See page F3, *Managing Transient Viremia, or 'Blips.'*

11th IAS Conference on HIV Science; July 18-21, 2021; Virtual

Presenting Author: Mark Underwood; mark.r.underwood@viivhealthcare.com

This content was acquired following an unsolicited medical information enquiry by a healthcare professional.

Always consult the product information for your country before prescribing a ViiV medicine. ViiV does not recommend the use of our medicines outside the terms of their licence.

In some cases, the scientific information requested and downloaded may relate to the use of our medicine(s) outside of their licence.